Safety, Pharmacokinetics, Pharmacodynamics, and Antitumor Activity of Necuparanib Combined with nab-Paclitaxel and Gemcitabine in Patients with Metastatic Pancreatic Cancer: Updated Phase 1 Results

Abstract #4117 / Poster #109

Eileen M. O'Reilly[1], Devalingam Mahalingam[2], James Roach[3], Paul Miller[3], Molly Rosano[3], Silva Krause[3], William Avery[3], Julie Wolf[4], Keith Flaherty[5], David Ryan[5] ; [1]Memorial Sloan Kettering Cancer Center, New York, NY; [2]Univ. of Texas Health Sciences Center, San Antonio, TX; [3]Momenta Pharmaceuticals, Cambridge, MA; [4]Novella Clinical, Boulder, CO; [5]Massachusetts General Hospital, Boston, MA

ABSTRACT

(Abstract as submitted in Feb. 2016)

BACKGROUND: Necuparanib ("necu") inhibits multiple heparin-binding growth factors, chemokines, and adhesion molecules in nonclinical studies. It is being evaluated in a Phase 1/2 trial with nab-paclitaxel (nabP) + gemcitabine (gem) in patients (pts) with metastatic pancreatic cancer. Updated Phase 1 safety/pharmacokinetic (PK)/pharmacodynamic (PD) data are provided.

METHODS: Necu was given as daily escalating s.c. doses in combination with i.v. 125 mg/m^2 nabP and 1000 mg/m^2 gem (Days 1, 8, 15 of each 28-day cycle). The necu start dose was 0.5 mg/kg and was escalated until the maximum tolerated dose was determined (which was 5 mg/kg). The protocol was amended to include nabP after 2 necu + gem cohorts were completed.

RESULTS: 39 pts in 7 cohorts (0.5 and 1 mg/kg + gem; 1, 2, 4, 6, and 5 mg/kg + nabP + gem) received necu. No new safety issues have been observed in this Phase 1 trial over the past year and no increase in known AEs of chemotherapy across doses was observed. Exposure to necu was observed starting at 2 mg/kg. Release of heparin-binding proteins (e.g. HGF, a PD marker) from heparan sulfate stores increased with dose and plateaued at 4-5 mg/kg. 3 (8%) pts remain alive; 19 (49%) pts have lived >1 year, 8 (21%) >1.5 years, and 4 (10%) >2 years; 33.7 months is the longest survival to date. 16 pts treated with necu + nabP + gem completed Cycle 1 and had ≥1 scan on treatment; 8 (50%) achieved RECIST partial response and 6 (38%) achieved stable disease, for a disease control rate of 14/16 (88%); median overall survival (OS) in this subset was 16.0 months. Median OS of patients treated with ≥1 dose of necu + nabP + gem (n=24) was 12.4 months. Of 15 CA19.9 evaluable pts, 15 (100%) had >20%, 14 (93%) had >50%, and 7 (47%) had >90% decreases from baseline.

CONCLUSIONS: Acceptable tolerability and encouraging signals of activity continue to be observed with the combination, which is being further evaluated in an ongoing randomized, double-blind, Phase 2 trial.

BACKGROUND

Reports of antitumor activity have been generated in clinical trials using heparins to treat venous thrombosis in cancer patients. As the anticoagulant activity of heparins is a major dose limiting factor, the potential of heparins in oncology has not yet been fully assessed. Necuparanib is a low anticoagulant, broad acting, glycol-split, heparan sulfate mimetic. The rationale behind developing necuparanib was to engineer a heparin-like molecule with substantially reduced anticoagulant activity that still retained potent activity against key molecules involved in tumor angiogenesis (e.g., VEGF, FGF-2, heparanase), progression (e.g., SDF-1α), and metastasis (e.g., P-selectin, heparanase). Necuparanib may offer advantages over existing heparin preparations in terms of increased anti-tumor efficacy and reduced risk of bleeding.

Necuparanib is currently in the Phase 2 portion of a Phase 1/2 trial (ClinicalTrials.gov NCT01621243) in patients with metastatic pancreatic cancer. **The objective of the current presentation is to summarize the Phase 1 results as of April 2016.**

METHODS

- **OBJECTIVES:** Safety/tolerability and Phase 2 dose determination (primary); PK/PD (secondary); antitumor activity, biomarkers (exploratory).
- **KEY SELECTION CRITERIA:** ≥18 yrs old; pancreatic ductal adenocarcinoma; metastasis documented by CT/MRI; ECOG 0 or 1; no prior radiotherapy/chemotherapy/surgery for adjuvant or metastatic pancreatic cancer.
- **STUDY TREATMENTS:** Necuparanib given as open-label daily escalating s.c. doses in combination with i.v. 125 mg/m^2 nab-paclitaxel (nabP; Abraxane®) and 1000 mg/m^2 gemcitabine (gem) on Days 1, 8, 15 of each 28-day cycle. The necuparanib start dose was 0.5 mg/kg, with escalation via a modified 3+3 design until the maximum tolerated dose (MTD) was determined (which was 5 mg/kg).

RESULTS

Baseline Characteristics / Dose Cohort Progression (N=39)

- Mean age: 63 yrs (min-max: 36-85)
- Sex: Female: 27/39 (69%); Male: 12/39 (31%)
- Race: White: 35/39 (90%); Black/African American: 2/39 (5%); Not reported: 2/39 (5%)
- Ethnicity: Hispanic/Latino: 6/39 (15%); Not Hispanic/Latino: 31/39 (80%); Not reported: 2/39 (5%)

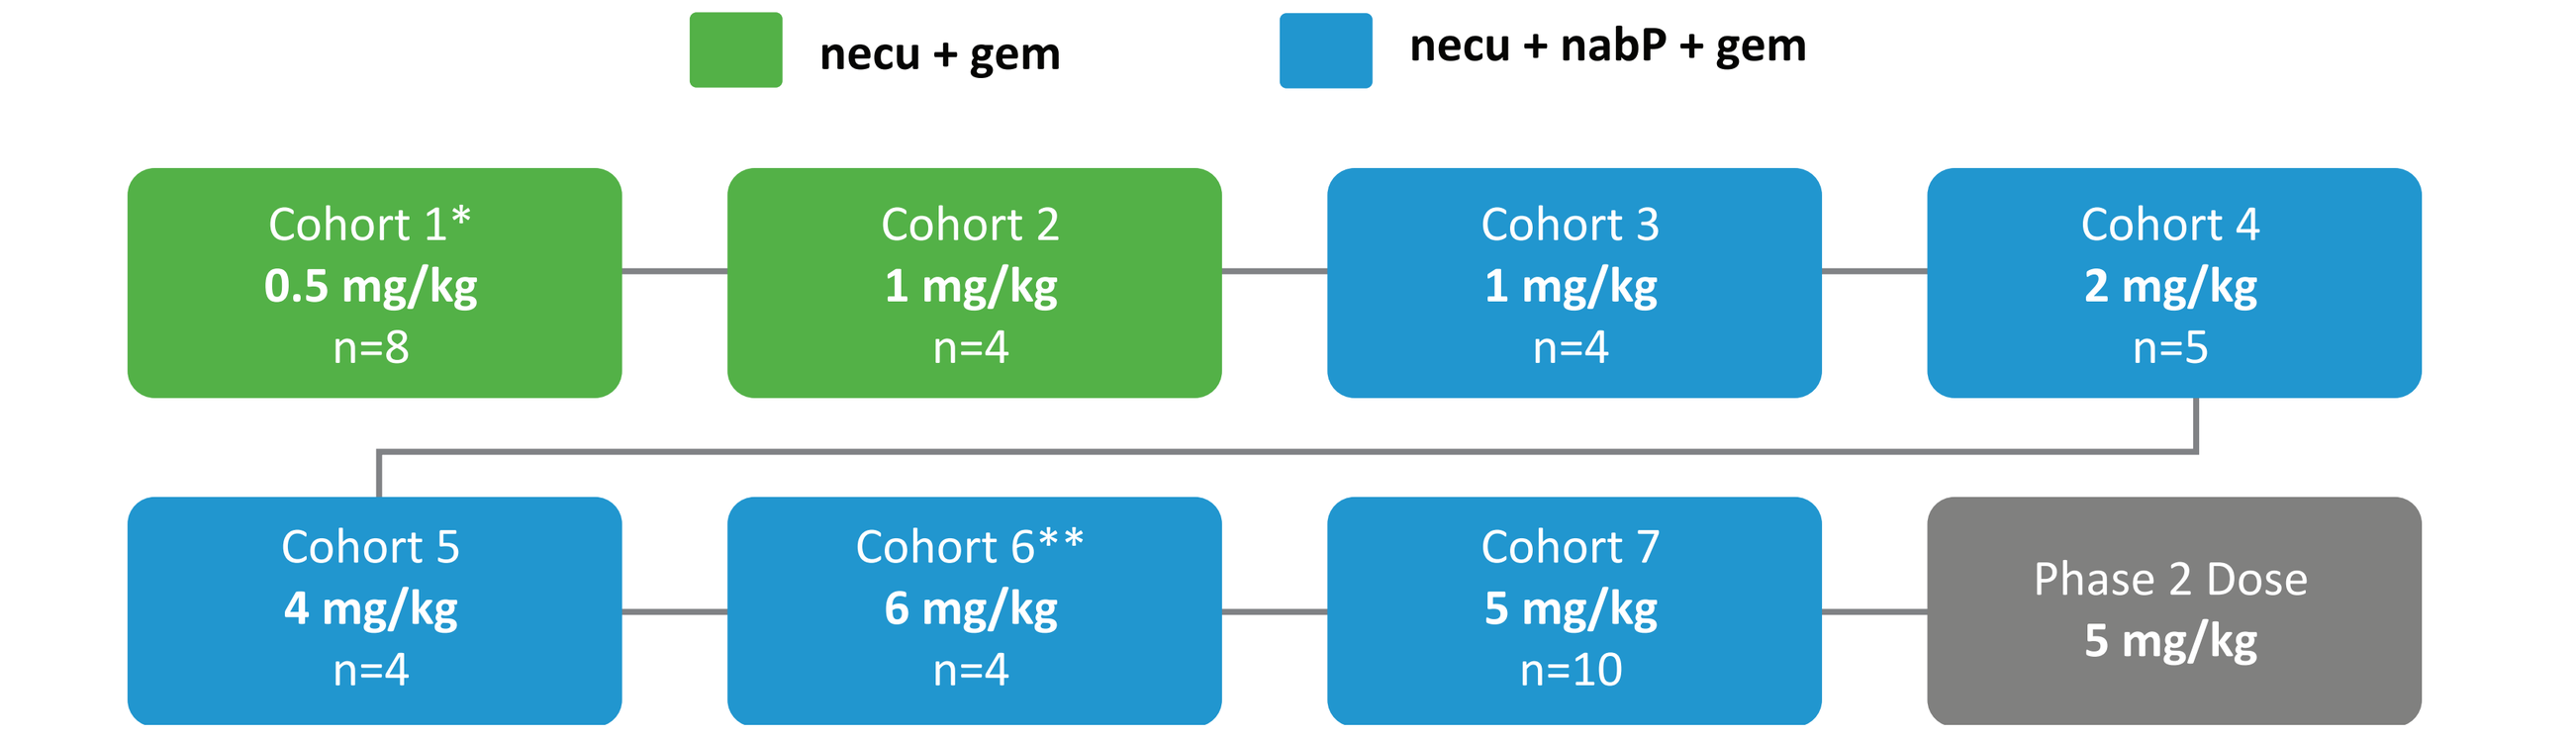

necu + gem
necu + nabP + gem
Cohort 1* 0.5 mg/kg n=8
Cohort 2 1 mg/kg n=4
Cohort 3 1 mg/kg n=4
Cohort 4 2 mg/kg n=5
Cohort 5 4 mg/kg n=4
Cohort 6** 6 mg/kg n=4
Cohort 7 5 mg/kg n=10
Phase 2 Dose 5 mg/kg

ns = pts receiving ≥1 dose necu, * dose-limiting toxicity (DLT) of ↑LFTs, ** DLT of cellulitis at injection site; also Grade 1 ↑aPTT

Most Common AEs (≥30% of Necu + nabP + Gem Patients)

Adverse Event	Co 1 0.5 mg/kg (n=8)	Co 2 1 mg/kg (n=4)	Co 1+2 TOTAL (n=12)	Co 3 1 mg/kg (n=4)	Co 4 2 mg/kg (n=5)	Co 5 4 mg/kg (n=4)	Co 6 6 mg/kg (n=4)	Co 7 5 mg/kg (n=10)	Co 3-7 TOTAL (n=27)
Anemia	5 (63)	3 (75)	**8 (67)**	3 (75)	4 (80)	2 (50)	2 (50)	3 (30)	**14 (52)**
Fatigue	4 (50)	2 (50)	**6 (50)**	2 (50)	3 (60)	3 (75)	2 (50)	4 (40)	**14 (52)**
Neutropenia	4 (50)	3 (75)	**7 (58)**	1 (25)	3 (60)	3 (75)	2 (50)	4 (40)	**13 (48)**
Leukopenia	3 (38)	1 (25)	**4 (33)**	2 (50)	3 (60)	2 (50)	3 (75)	2 (20)	**12 (44)**
Thrombocytopenia	2 (25)	2 (50)	**4 (33)**	2 (50)	2 (40)	4 (100)	2 (50)	2 (20)	**12 (44)**
ALT ↑	1 (13)	1 (25)	**2 (17)**	2 (50)	2 (40)	3 (75)	3 (75)	1 (10)	**11 (41)**
Nausea	1 (13)	2 (50)	**3 (25)**	3 (75)	3 (60)	2 (50)	1 (25)	2 (20)	**11 (41)**
Abdominal pain	1 (13)	-	**1 (8)**	3 (75)	1 (20)	2 (50)	1 (25)	3 (30)	**10 (37)**
Diarrhea	2 (25)	2 (50)	**4 (33)**	1 (25)	2 (40)	3 (75)	2 (50)	1 (10)	**9 (33)**
AST ↑	1 (13)	1 (25)	**2 (17)**	2 (50)	2 (40)	1 (25)	2 (50)	1 (10)	**8 (30)**
Hyperglycemia	3 (38)	-	**3 (25)**	-	2 (40)	2 (50)	3 (75)	1 (10)	**8 (30)**
Hypoalbuminemia	-	-	**-**	1 (25)	2 (40)	1 (25)	3 (75)	1 (10)	**8 (30)**
Peripheral neuropathy	-	-	**-**	1 (25)	1 (20)	2 (50)	1 (25)	3 (30)	**8 (30)**

n (%) patients are shown. "-" = no AEs. Co 1& 2 = necu +gem; Co 3-7 = necu + nabP + gem.

AEs Grade 3 or Higher, Probably/Definitely Related to Necu: >1 Necu + nabP + Gem Patient

Adverse Event	Co 1 0.5 mg/kg (n=8)	Co 2 1 mg/kg (n=4)	Co 1+2 TOTAL (n=12)	Co 3 1 mg/kg (n=4)	Co 4 2 mg/kg (n=5)	Co 5 4 mg/kg (n=4)	Co 6 6 mg/kg (n=4)	Co 7 5 mg/kg (n=10)	Co 3-7 TOTAL (n=27)
ALT ↑	1 (13)	1 (25)	**2 (17)**	1 (25)	1 (20)	1 (25)	1 (25)	-	**4 (15)**
Anemia	-	-	**-**	-	-	-	1 (25)	-	**1 (4)**
Blood alk phos ↑	-	-	**-**	-	-	-	-	1 (10)	**1 (4)**
AST ↑	1 (13)	-	**1 (8)**	-	-	-	1 (25)	-	**1 (4)**
Inj site cellulitis	-	-	**-**	-	-	-	1 (25)	-	**1 (4)**
Lymphopenia	-	-	**-**	-	-	1 (25)	-	-	**1 (4)**
Neutropenia	-	-	**-**	-	1 (20)	-	-	-	**1 (4)**

n (%) patients are shown. "-" = no AEs. Co 1& 2 = necu +gem; Co 3-7 = necu + nabP + gem.

Patient Time on Study: Cohorts 1-2 (Necu + Gem)

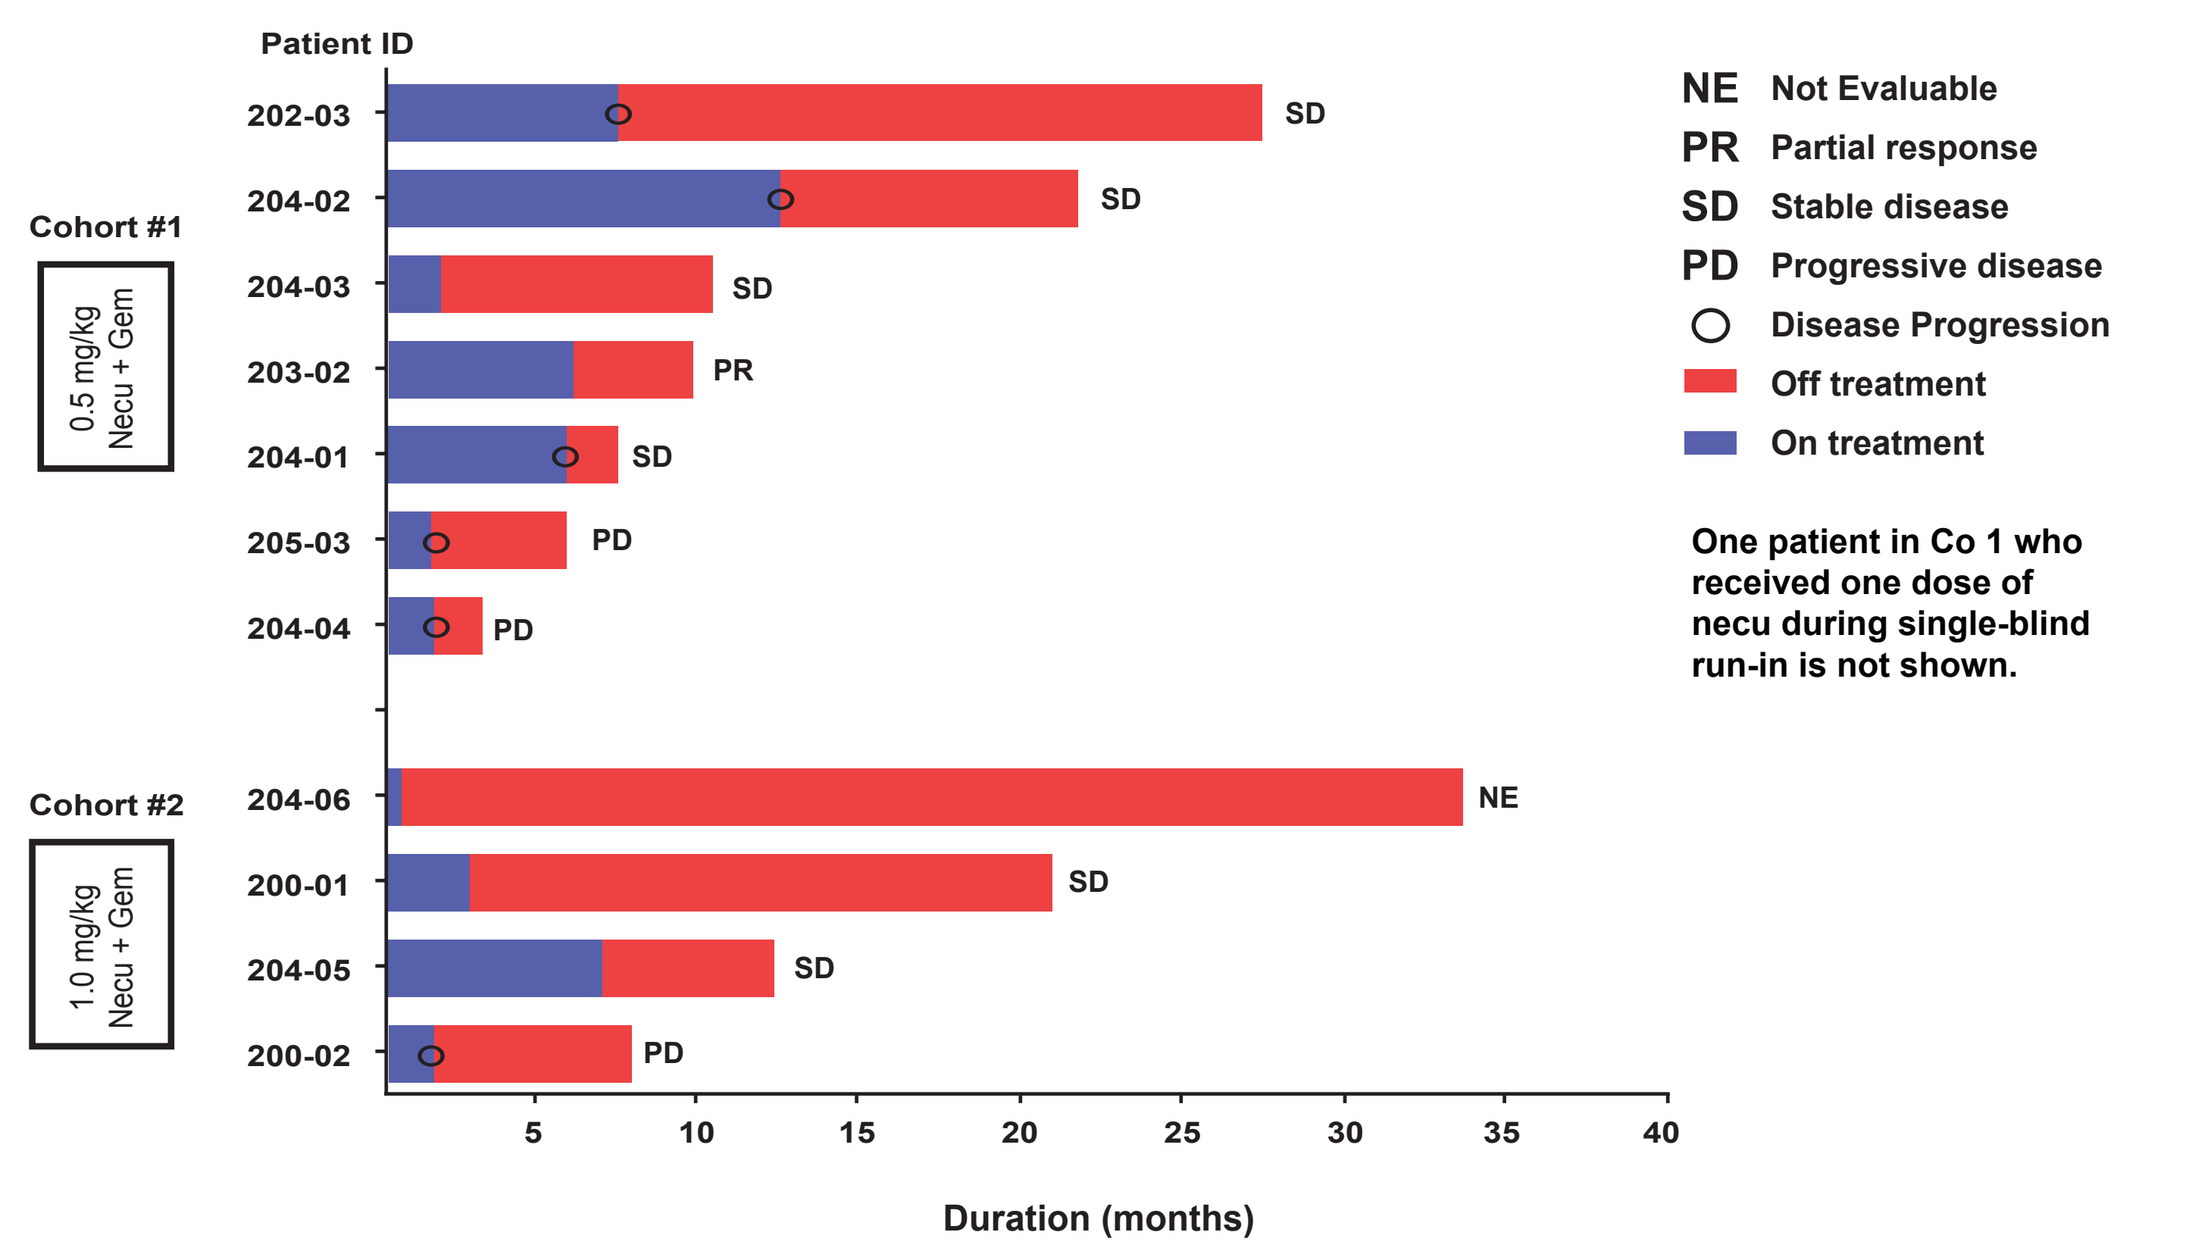

Patient ID
Cohort #1
0.5 mg/kg Necu + Gem
Cohort #2
1.0 mg/kg Necu + Gem
Duration (months)
NE Not Evaluable
PR Partial response
SD Stable disease
PD Progressive disease
Disease Progression
Off treatment
On treatment
One patient in Co 1 who received one dose of necu during single-blind run-in is not shown.

Patient Time on Study: Cohorts 3-7 (Necu + nabP + Gem)

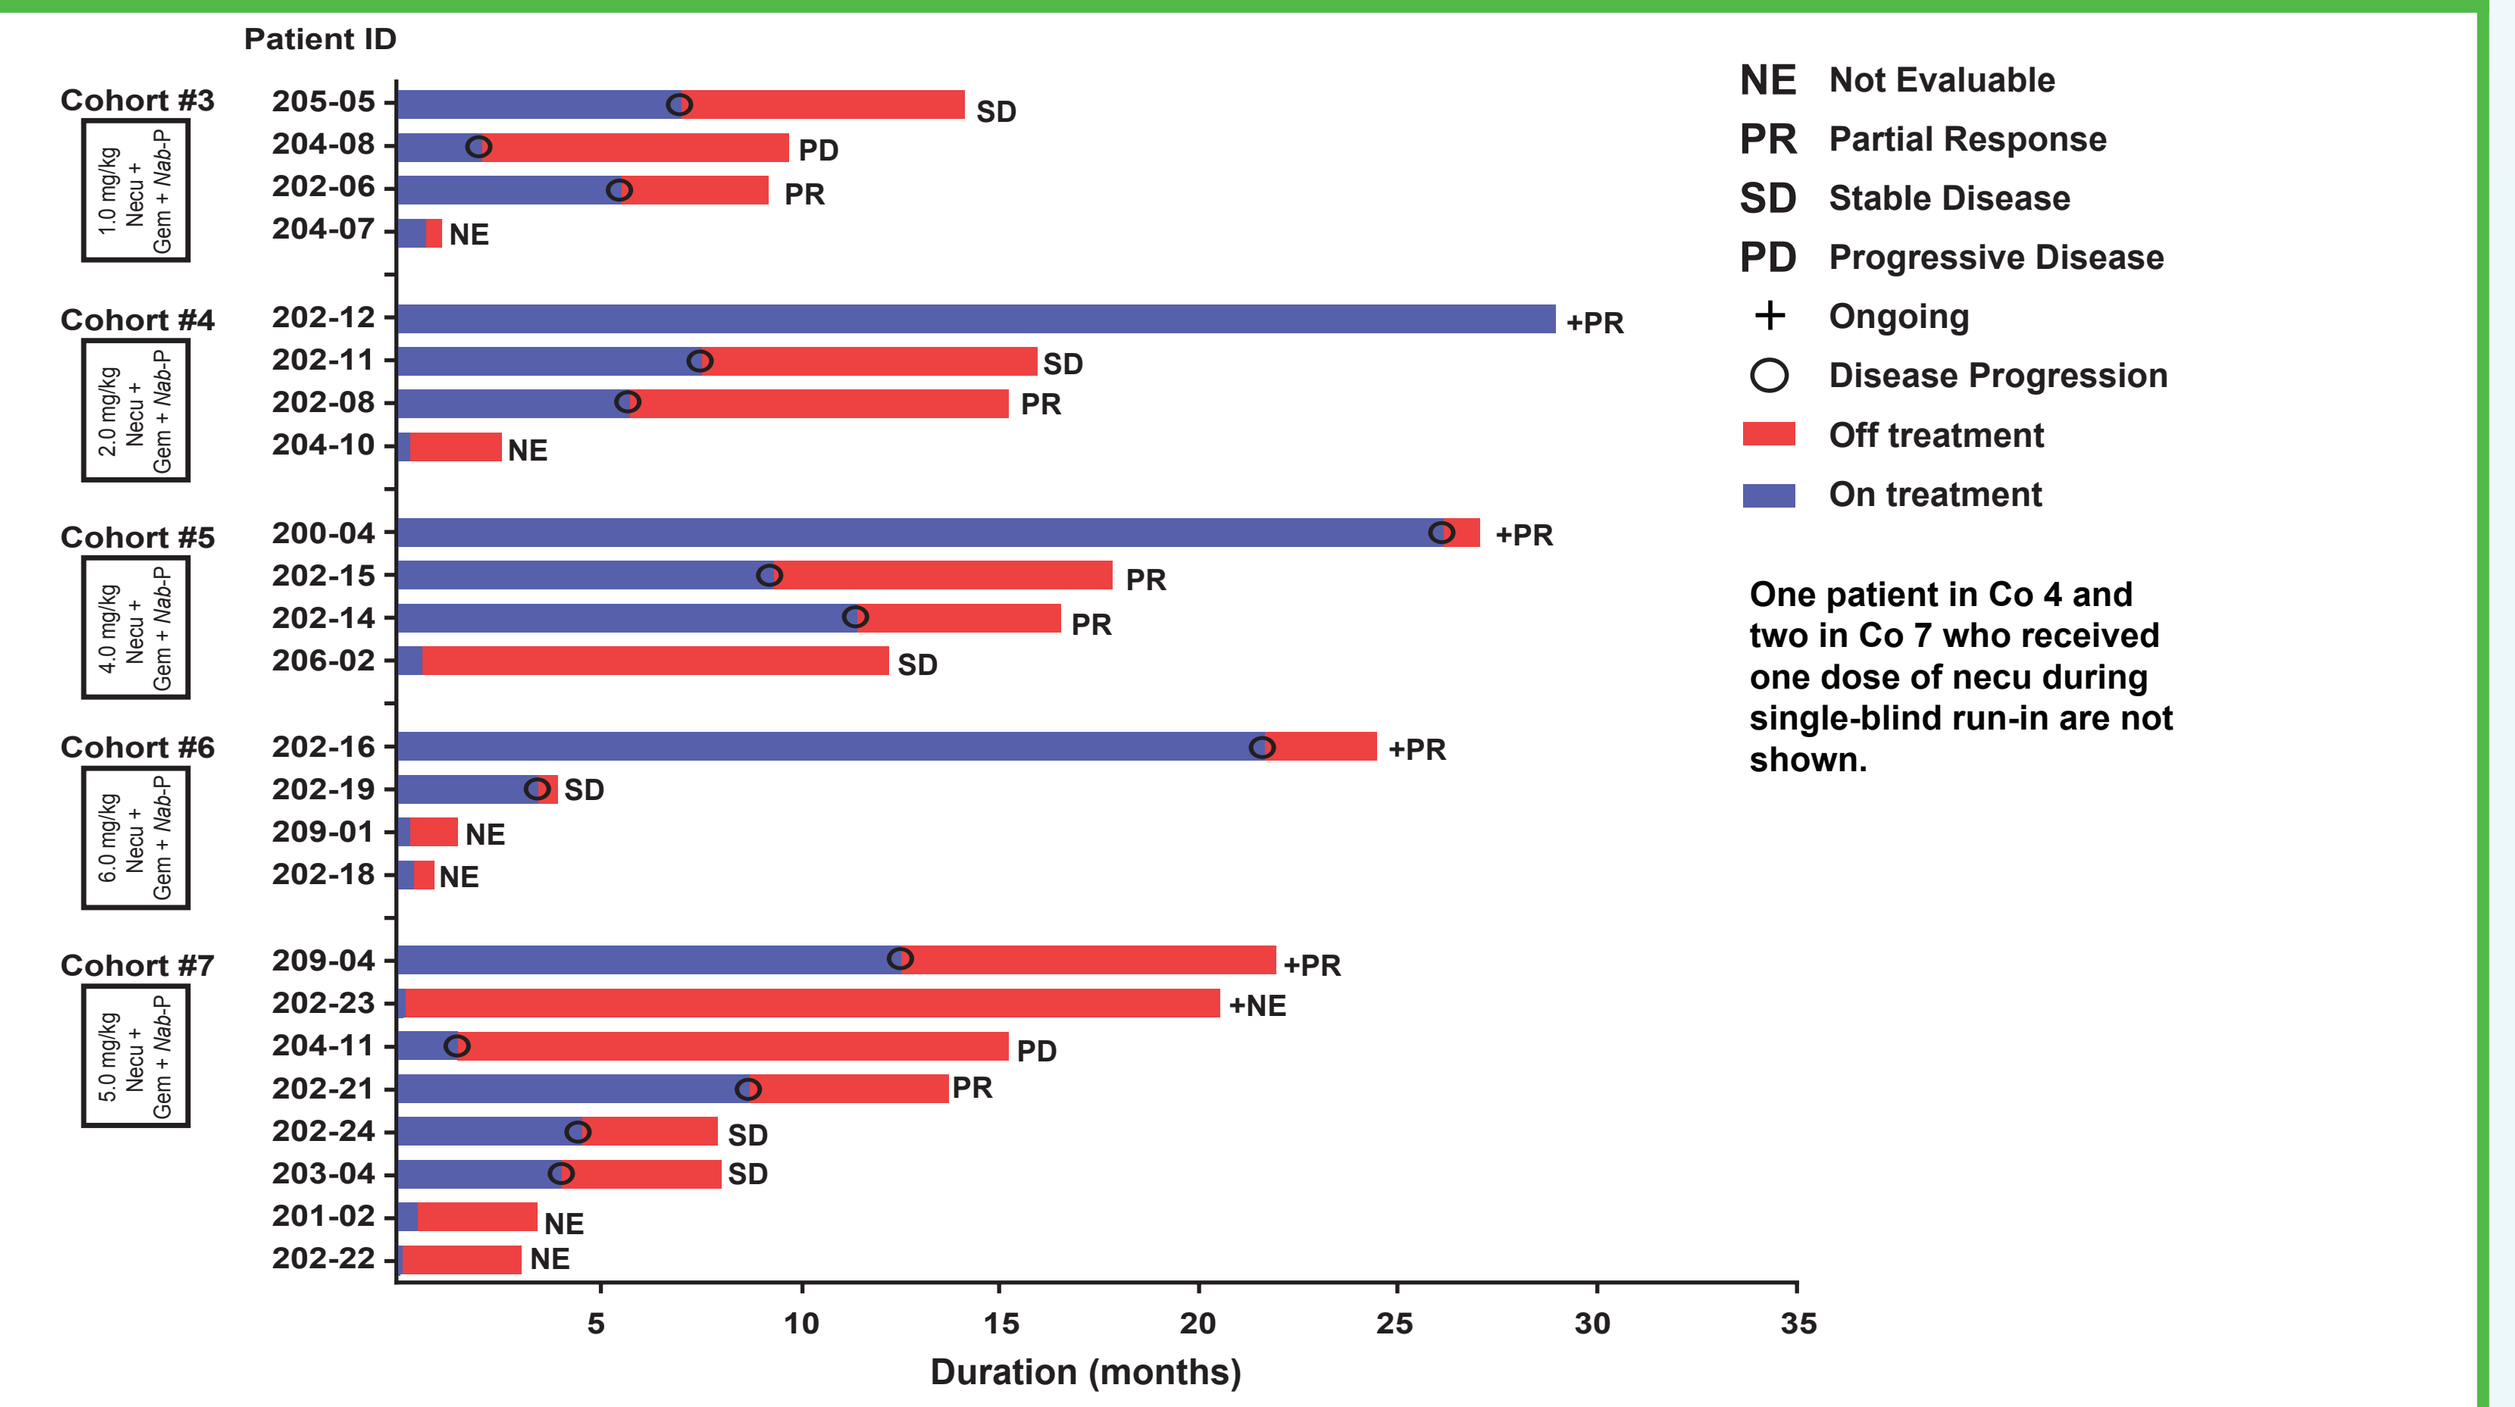

Patient ID
Cohort #3
Cohort #4
Cohort #5
Cohort #6
Cohort #7
Duration (months)
NE Not Evaluable
PR Partial Response
SD Stable Disease
PD Progressive Disease
+ Ongoing
Disease Progression
Off treatment
On treatment
One patient in Co 4 and two in Co 7 who received one dose of necu during single-blind run-in are not shown.

Maximum Change in RECIST Target Lesion Size

During the past year, 4 patients had larger decreases and 1 changed from SD to PR

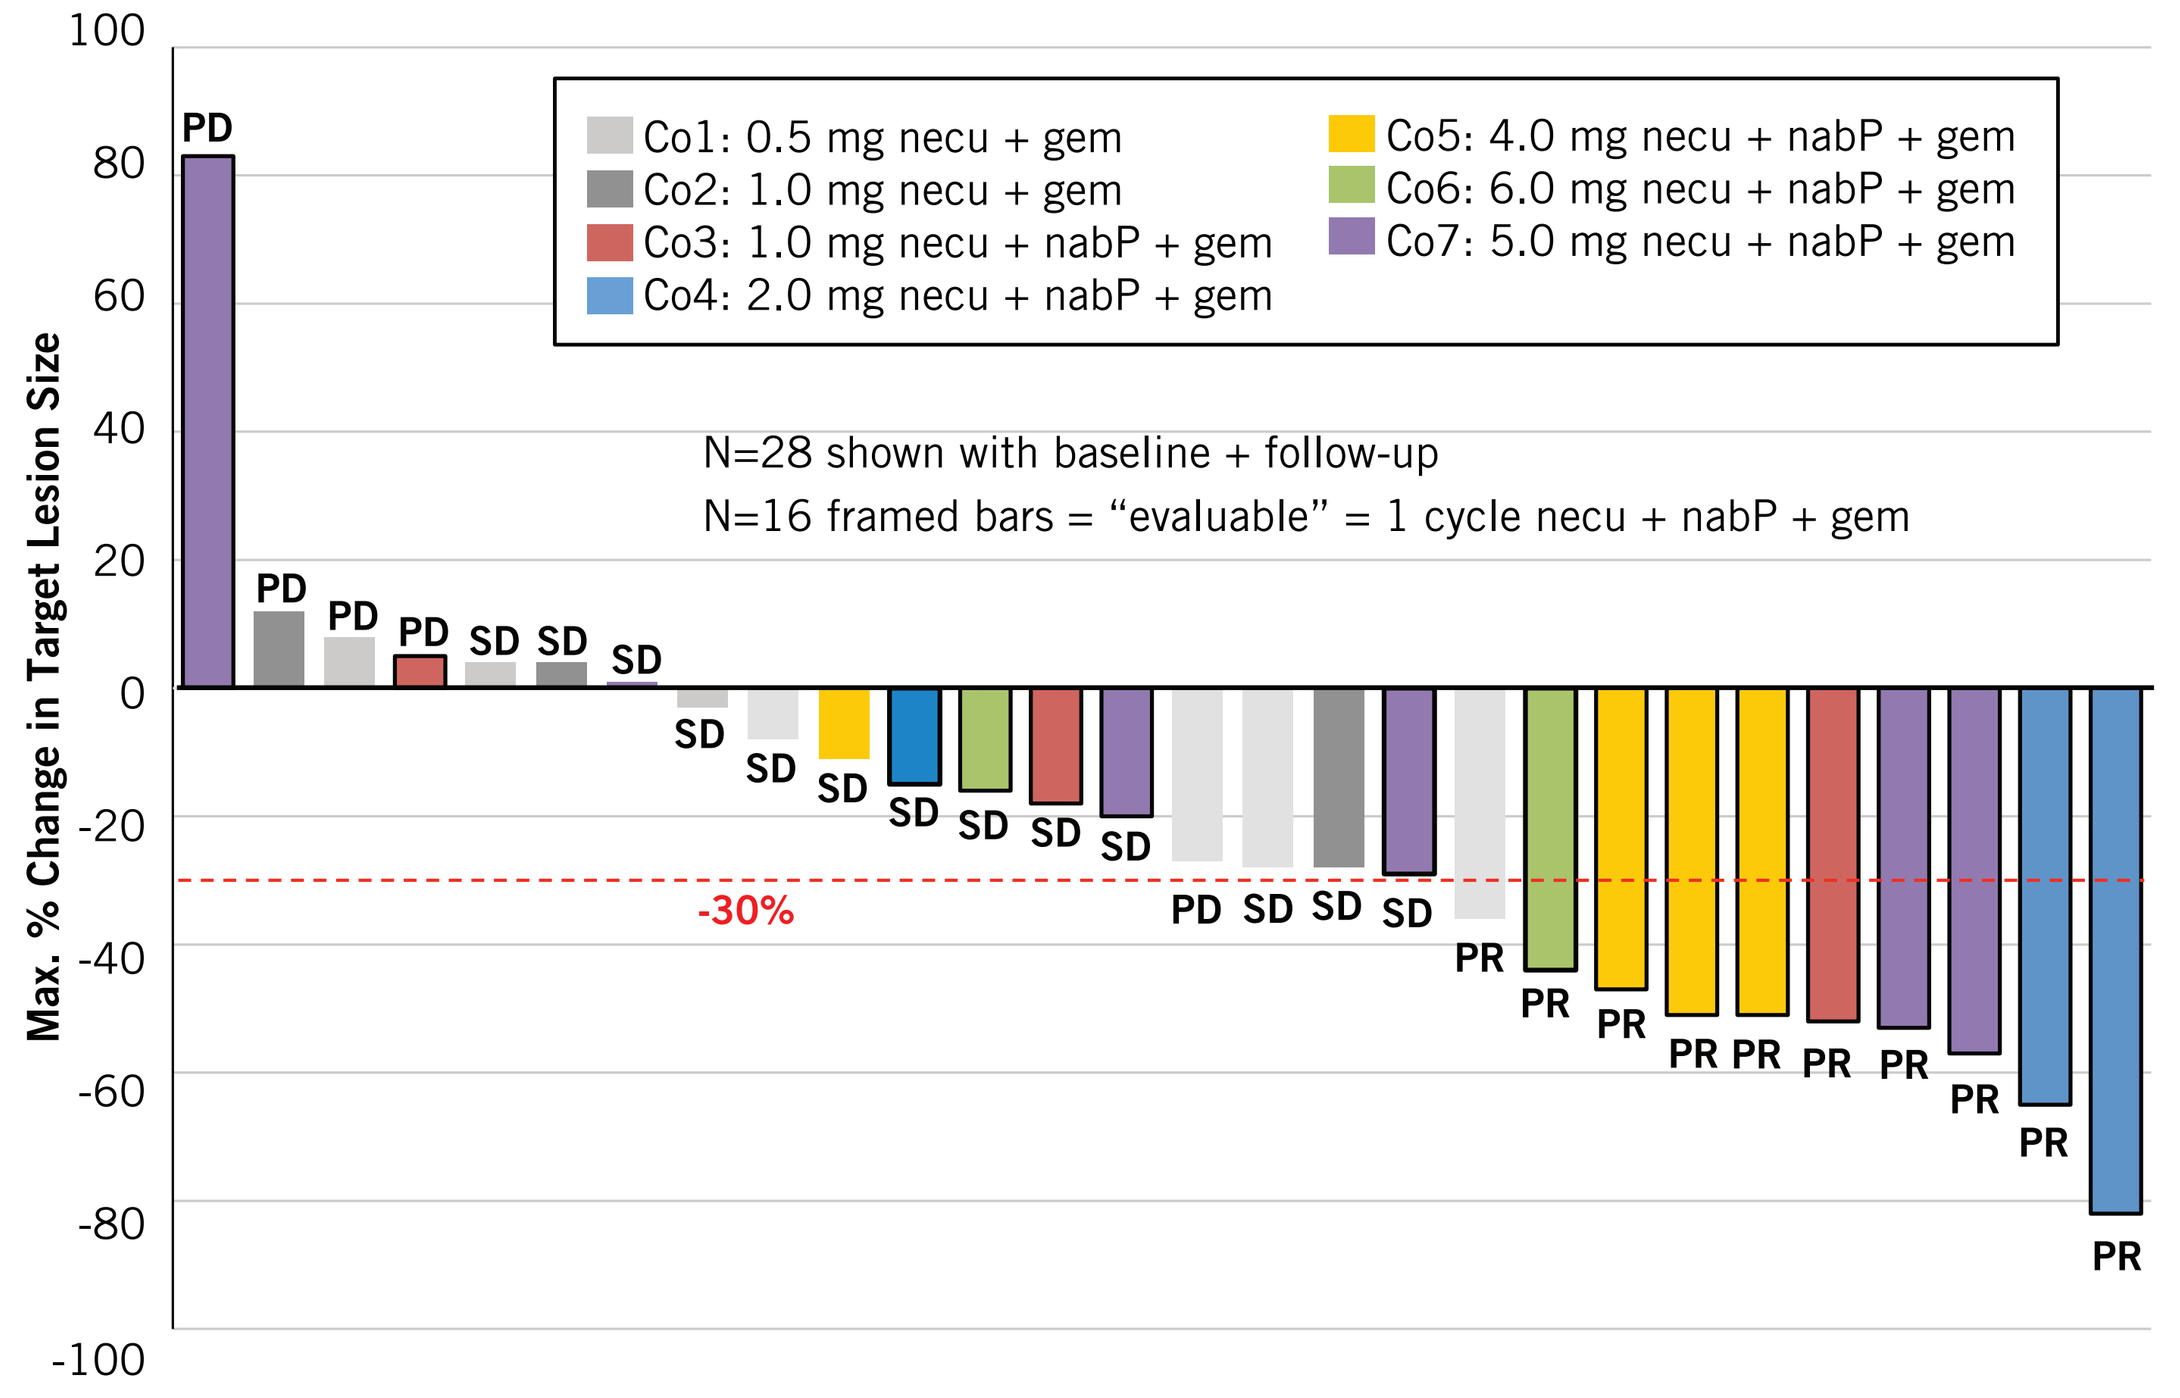

Max. % Change in Target Lesion Size
Co1: 0.5 mg necu + gem
Co2: 1.0 mg necu + gem
Co3: 1.0 mg necu + nabP + gem
Co4: 2.0 mg necu + nabP + gem
Co5: 4.0 mg necu + nabP + gem
Co6: 6.0 mg necu + nabP + gem
Co7: 5.0 mg necu + nabP + gem
N=28 shown with baseline + follow-up
N=16 framed bars = "evaluable" = 1 cycle necu + nabP + gem
-30%

Maximum Change in CA19.9 Levels

During the past year, 1 patient had a larger decrease and changed from SD to PR

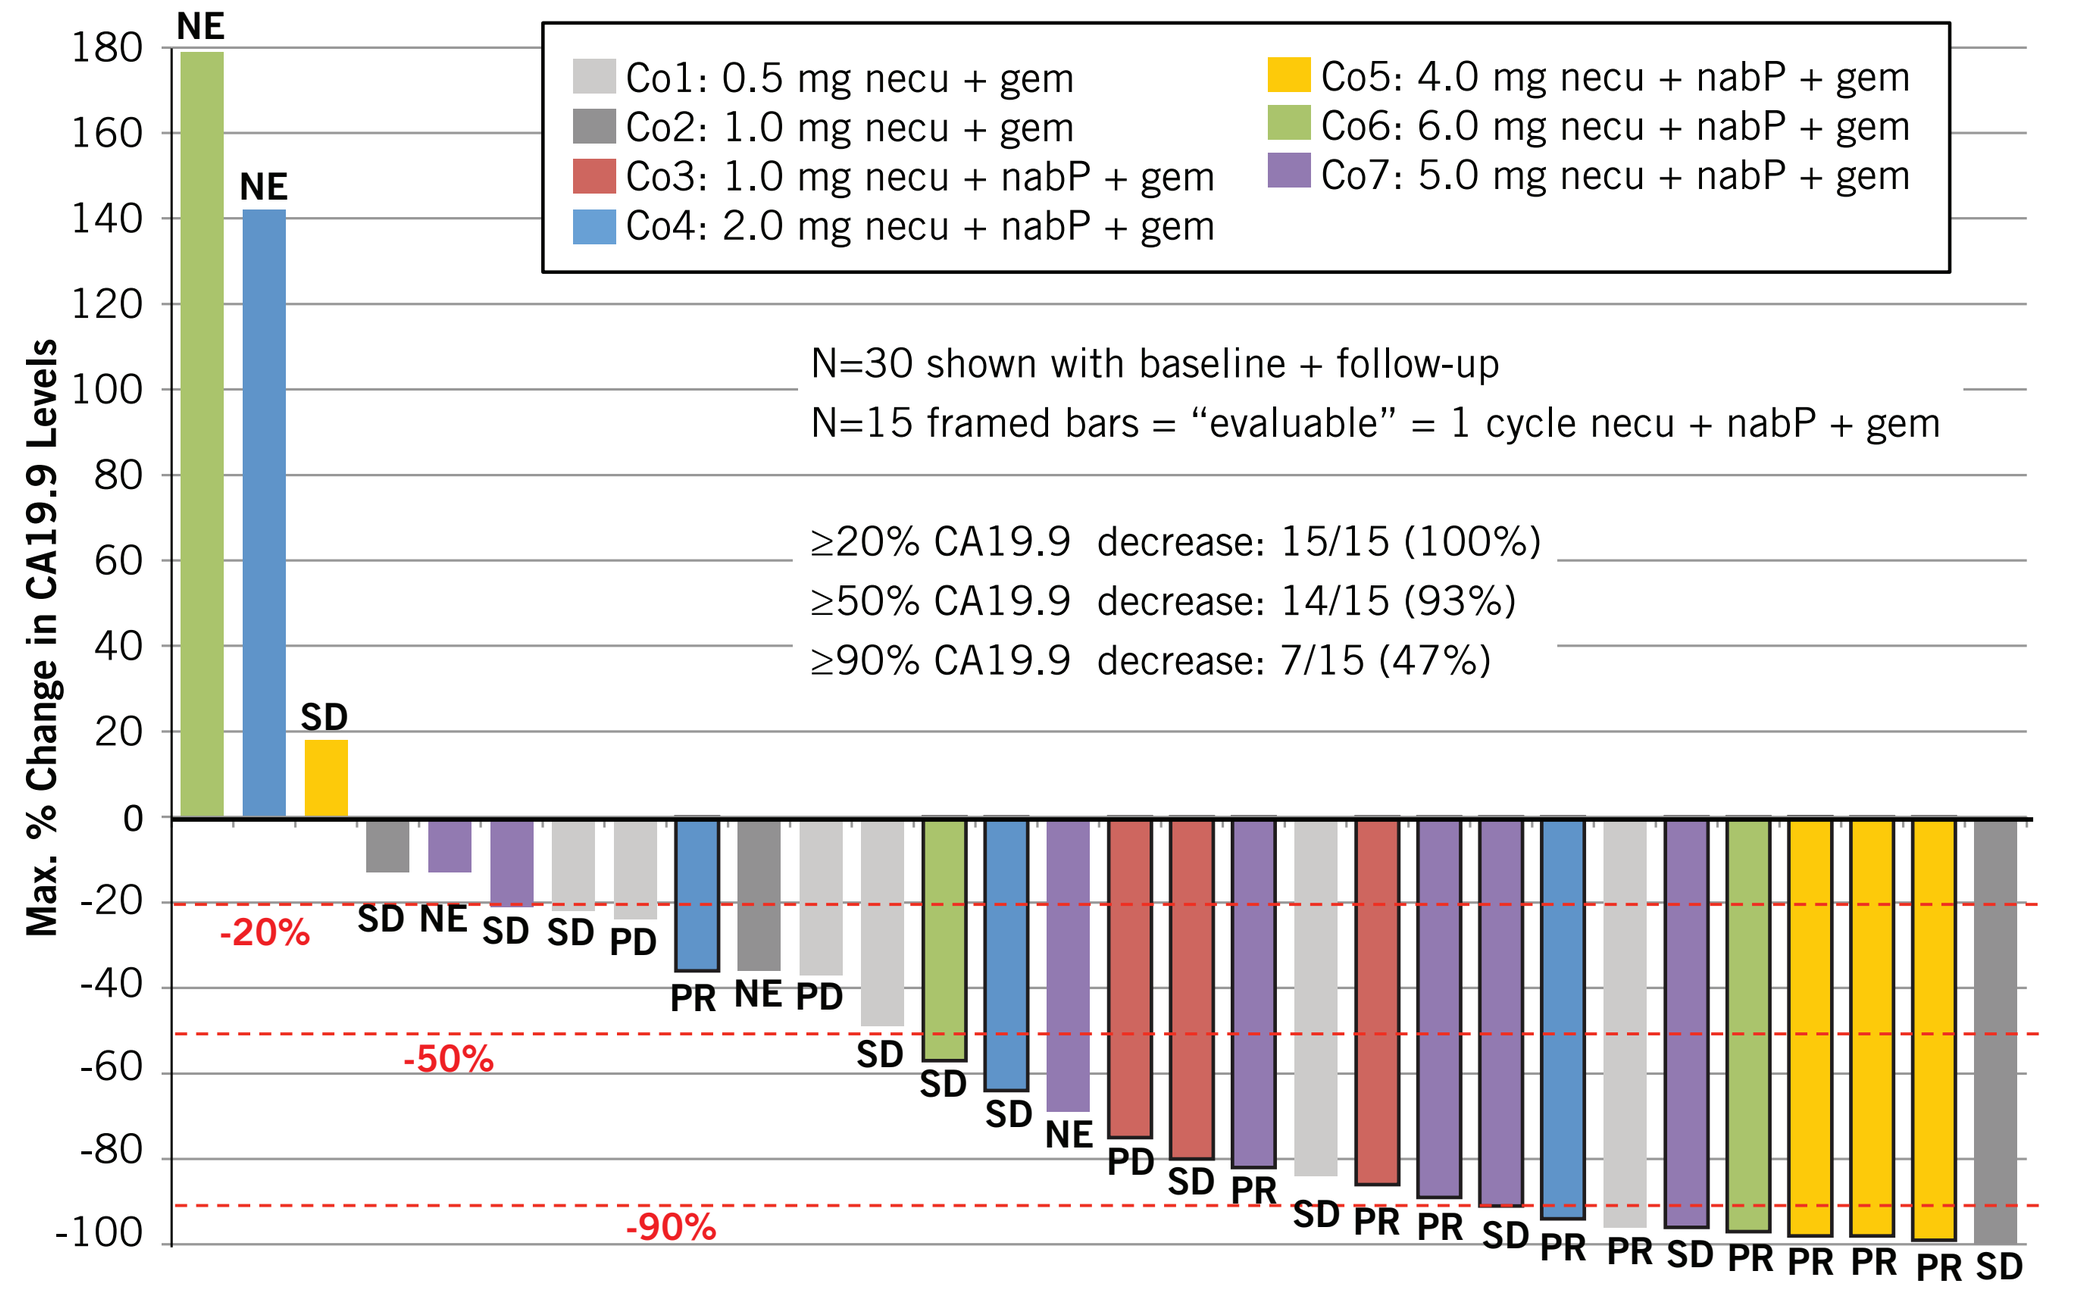

Max. % Change in CA19.9 Levels
Co1: 0.5 mg necu + gem
Co2: 1.0 mg necu + gem
Co3: 1.0 mg necu + nabP + gem
Co4: 2.0 mg necu + nabP + gem
Co5: 4.0 mg necu + nabP + gem
Co6: 6.0 mg necu + nabP + gem
Co7: 5.0 mg necu + nabP + gem
N=30 shown with baseline + follow-up
N=15 framed bars = "evaluable" = 1 cycle necu + nabP + gem
≥20% CA19.9 decrease: 15/15 (100%)
≥50% CA19.9 decrease: 14/15 (93%)
≥90% CA19.9 decrease: 7/15 (47%)
-20%
-50%
-90%

Pharmacokinetics/Pharmacodynamics

- PK data are shown for patients with at least 3 measurable levels of necuparanib detected at the time points assessed during Day 1. Measurable levels of necuparanib were seen starting at the 2 mg/kg dose group. Long-term stability testing of the PK samples is ongoing and the data represented here should be considered preliminary.

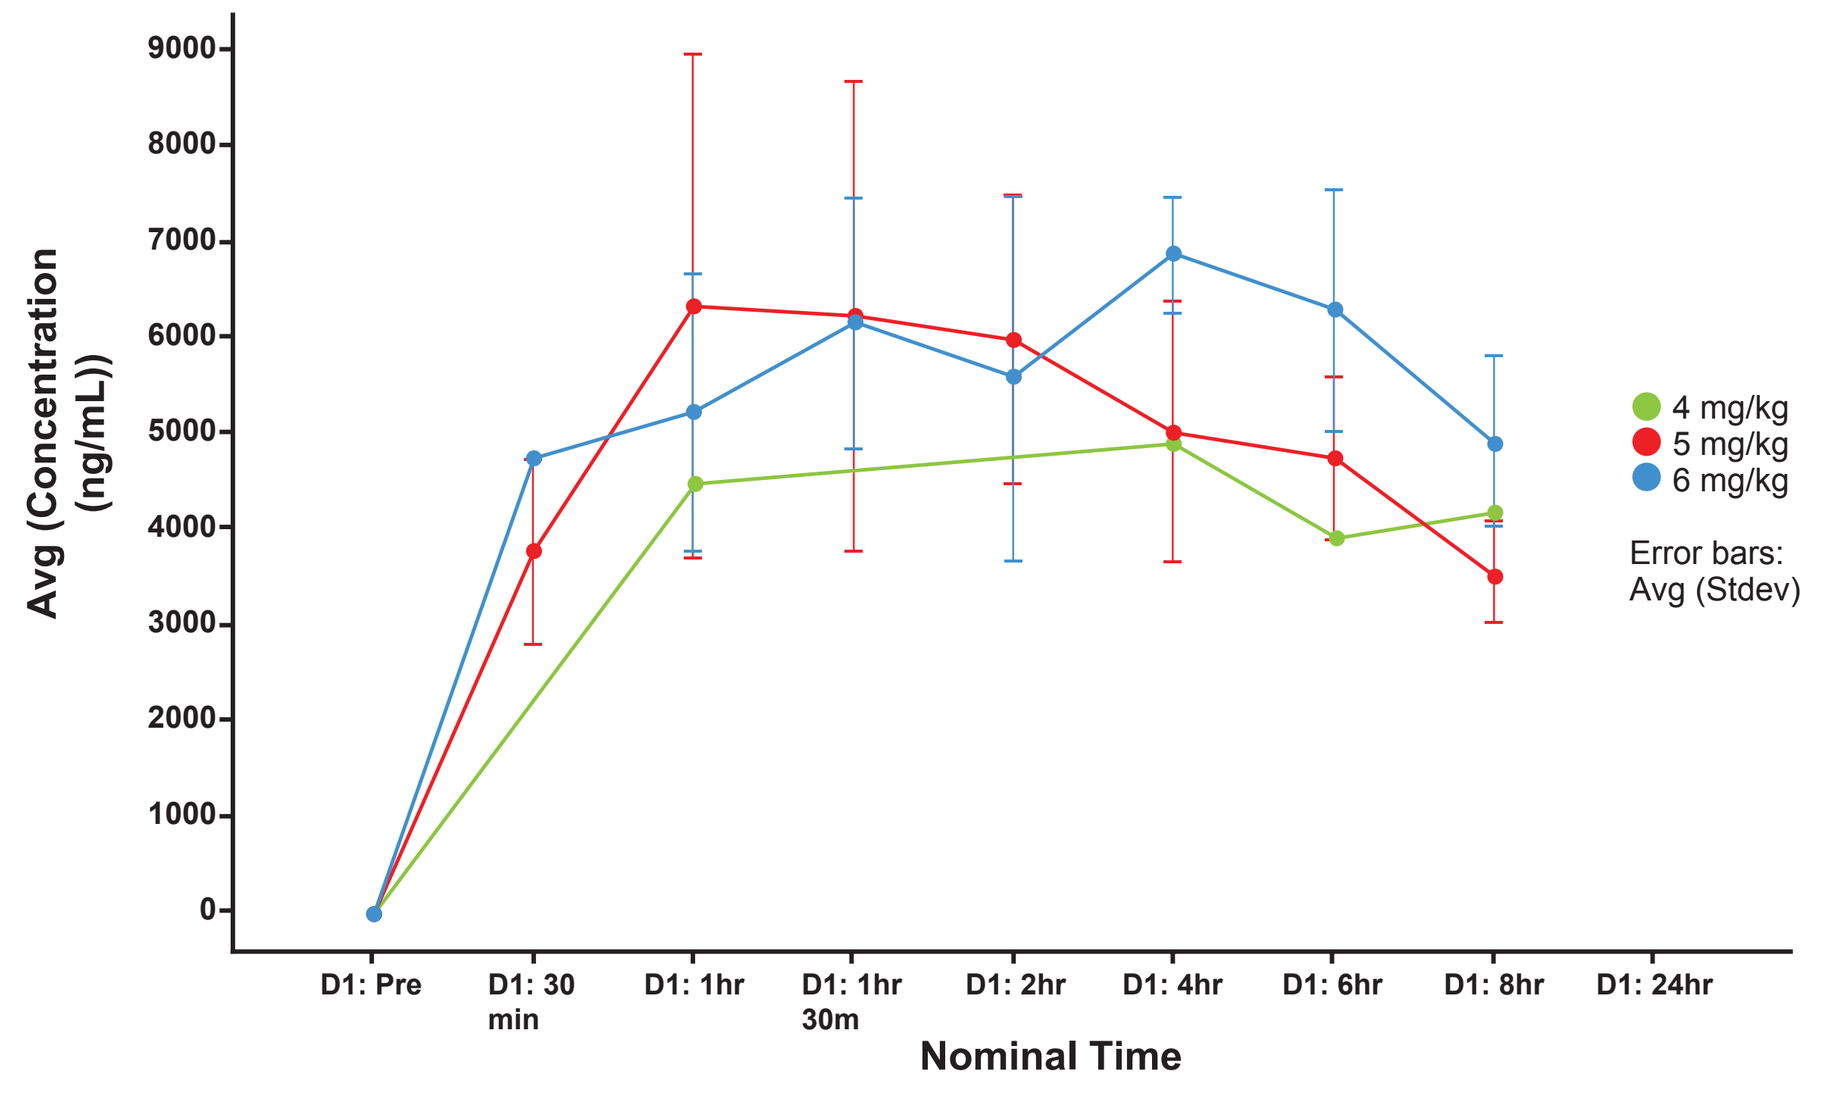

Avg (Concentration (ng/mL))
Nominal Time
D1: Pre
D1: 30 min
D1: 1hr
D1: 1hr 30m
D1: 2hr
D1: 4hr
D1: 6hr
D1: 8hr
D1: 24hr
4 mg/kg
5 mg/kg
6 mg/kg
Error bars: Avg (Stdev)

- Release of heparin-binding proteins (e.g. HGF, a PD marker) from heparan sulfate stores increased with dose and plateaued at 4-5 mg/kg.

OS/PFS/Response Data

Efficacy Variable	Necu + gem ≥1 Dose (n=11)	Necu + gem ≥1 Cycle (n=10)	Necu + nabP + gem ≥1 Dose (n=24)	Necu + nabP + gem ≥1 Cycle (n=16)
OS (mo) median (95% CI)	10.4 (6.1-21.8)	10.2 (3.4-21.0)	13.1 (4.0-16.6)	15.6 (9.3-17.8)
Survival Rate % (95% CI)				
6 months	91 (51-99)	90 (47-99)	71 (48-85)	94 (63-99)
12 months	45 (17-71)	40 (12-67)	54 (33-71)	69 (40-86)
18 months	36 (11-63)	30 (7-58)	21 (8-39)	25 (8-47)
24 months	18 (3-44)	10 (1-36)	21 (8-39)	25 (8-47)
PFS (mo) median (95% CI)	7.5 (1.9-12.5)	6.5 (1.6-10.4)	5.9 (2.1-8.7)	7.9 (3.4-11.4)
RECIST Response (n, %)				
Best response – unconfirmed or confirmed				
CR	0	0	0	0
PR	1 (9)	1 (10)	9 (38)	9 (56)
SD	6 (55)	6 (60)	6 (25)	5 (31)
PD	3 (27)	3 (30)	2 (8)	2 (13)
NE	1 (9)	0	7 (29)	0
Disease Control Rate	7 (64)	7 (70)	15 (63)	14 (88)

OS=overall survival; PFS=progression-free survival; CR=complete response; PR=partial response; SD=stable disease; PD=progressive disease; NE=not evaluable; disease control rate=CR+PR+SD.

SUMMARY

- No increases in incidence, severity, or duration for known AEs of gem or nabP + gem were observed when combined with necuparanib.
- Hematological toxicities were monitored, with the following overall rates of Grade 3 or 4 AEs in necu + nabP + gem patients: neutropenia (37%), anemia (22%), leukopenia (19%), thrombocytopenia (19%), lymphopenia (11%), and no Grade 3/4 AEs of leukocytosis, febrile neutropenia, epistaxis, pulmonary embolism, DVT, phlebitis, or hematuria. With the exception of anemia, these rates were similar to or lower than those observed in the Von Hoff Phase 1/2 study (neutropenia, 75%; anemia, 14%; thrombocytopenia, 28%; and leukopenia, 56%) [3].
- Measurable levels of necuparanib were seen starting at the 2 mg/kg dose group. Release of HGF increased with increasing doses and plateaued at 4-5 mg/kg.
- Encouraging signals of activity were observed:
 - 16 patients treated with necu + nabP + gem completed Cycle 1 and had ≥1 scan on treatment; 9 (56%) achieved RECIST partial response (PR) and 5 (31%) achieved stable disease, for a disease control rate (DCR) of 14/16 (88%); median OS in this subset was 15.6 months. Median OS of patients treated with ≥1 dose of necu + nabP + gem (n=24) was 13.1 months.
 - 24-month survival rates: 25% in patients who received ≥1 cycle and 21% in patients who received ≥1 dose.
 - Of 15 CA19.9 evaluable patients, 15 (100%) had ≥20%, 14 (93%) had ≥50%, and 7 (47%) had ≥90% decreases from baseline.
 - Noting differences in sample sizes and study populations, these results compared favorably with published nabP + gem data:
 - Phase 1/2 data: 48% PR; 68% DCR; 12.2 months OS; 78% patients had ≥50% CA19.9 decrease [3].
 - Phase 3 data: 23% PR; 50.3% DCR; 8.5 months OS; 9% 24-month survival rate [4].

CONCLUSIONS

- Acceptable tolerability and encouraging signals of activity continue to be observed with the combination, which is being further evaluated in an ongoing randomized, double-blind, Phase 2 trial.

REFERENCES

1. Zhou et al. M402, a novel heparan sulfate mimetic, targets multiple pathways implicated in tumor progression and metastasis. PLoS ONE 2011;6(6):e21106.
2. Schultes et al. M402, a Heparan Sulfate Mimetic and Novel Candidate for the Treatment of Pancreatic Cancer. Poster presented at ASCO Annual Meeting 2012.
3. Von Hoff, et al. Gemcitabine plus nab-paclitaxel is an active regimen in patients with advanced pancreatic cancer: a phase I/II trial. J Clin Onc 2011;29(34):4548.
4. Von Hoff et al. Increased Survival in Pancreatic Cancer with nab-Paclitaxel plus Gemcitabine NEJM 2013;369(18):1691-703.

Presented at the 2016 ASCO Annual Meeting
June 4, 2016
Chicago, IL, USA

MOMENTA

© 2016 Momenta Pharmaceuticals, Inc. All rights reserved.