M402, a Novel Drug Candidate for Pancreatic Cancer: Efficacy in Animal Models and Initial Clinical Study Design

David Ryan[1], Keith Flaherty[1], David Tuveson[2], Martijn Lolkema[3], Birgit Schultes[4], Chia Lin Chu[4], Alison Long[4], Paul Miller[4], Molly Rosano[4], Michelle Cyr[4], James Roach[4]

[1]Massachusetts General Hospital, Boston, MA; [2]Cold Spring Harbor Laboratory, NY; [3]Univ. Med. Center, Utrecht, Netherlands; [4]Momenta Pharmaceuticals, Cambridge, MA

Background

- M402's development is based on nonclinical studies showing heparins impacting pathways related to the pathogenesis of cancer as well as decades of reports of antitumor activity of heparins in cancer patients. Because the dose of these medicines has been limited by their anticoagulant activity, M402 – derived from unfractionated heparin – has been designed to have significantly reduced anticoagulant activity while optimizing heparin's antitumor properties.
- Nonclinical data showed potent binding of M402 to multiple growth factors, adhesion molecules, and chemokines to inhibit tumor progression, metastasis, and angiogenesis. Results from two models of pancreatic cancer are presented.

Methods

- **Genetic Model**: The Kras$^{G12D/+}$p53$^{R172H/flox}$PdxCre transgenic mouse spontaneously develops pancreatic tumors by Day 40. Metastasis to LN, liver, spleen, and lung. Considered one of the most physiologically relevant models of pancreatic cancer. Starting on Day 30: M402 or saline (SAL) via minipump, or gemcitabine (GEM) 50 mg/kg, q3d or SAL.
- **Capan-2** cells implanted into pancreas on Day 0; M402 40 mg/kg/day via osmotic pump starting on Wk 1 or 5; GEM at 30, 45, 60 mg/kg twice weekly starting on Day 28.

Results

- **Genetic Model Results**: M402 + GEM offered a survival benefit over GEM (right) and reduced the metastatic rate (below).

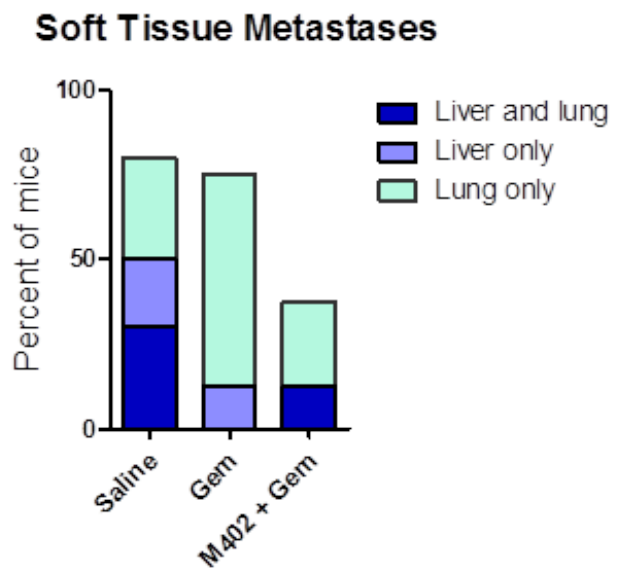
Soft Tissue Metastases
Percent of mice
100
50
0
Liver and lung
Liver only
Lung only
Saline
Gem
M402 + Gem

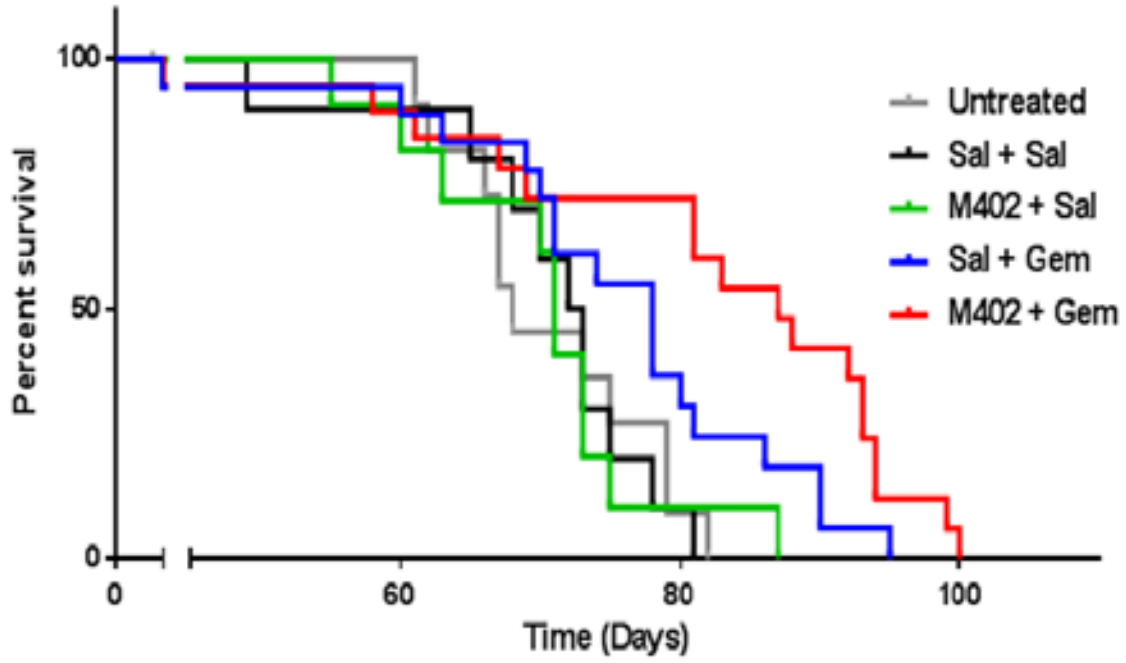
Percent survival
100
50
0
0
60
80
100
Time (Days)
Untreated
Sal + Sal
M402 + Sal
Sal + Gem
M402 + Gem

- **Capan-2 Results:** M402 reduced tumor growth as monotherapy and when combined with GEM (below).

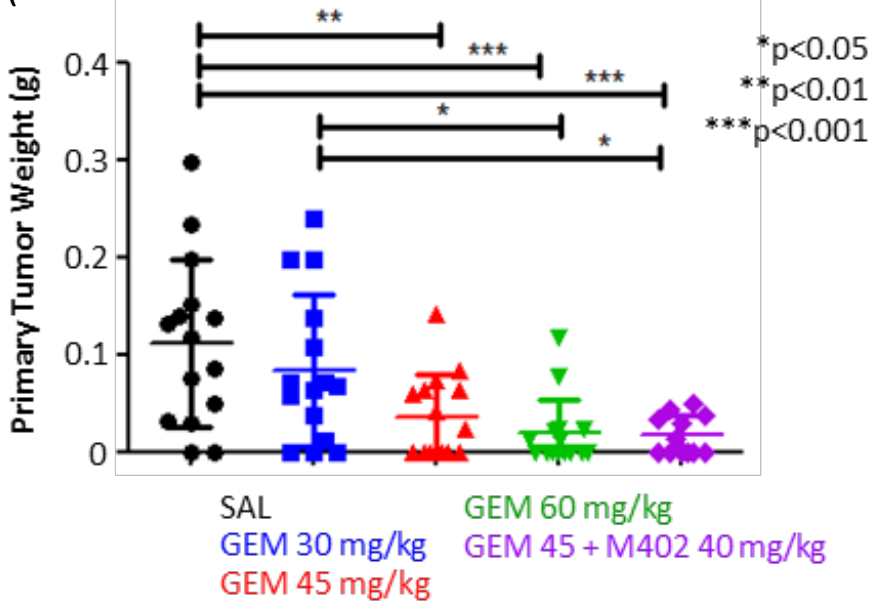
Primary Tumor Weight (g)
0.4
0.3
0.2
0.1
0
**

*
*
*p<0.05
**p<0.01
***p<0.001
SAL
GEM 30 mg/kg
GEM 45 mg/kg
GEM 60 mg/kg
GEM 45 + M402 40 mg/kg

Phase 1/2 Study

- Part A is a first-in-human, open-label, multiple ascending dose study in patients with metastatic pancreatic cancer. Part A objectives: to evaluate the safety and tolerability of M402 in combination with Abraxane® (nab-paclitaxel; nabP) and GEM and to establish the dose of M402 to take forward in Part B.
- Part B will be a randomized, controlled study investigating the antitumor activity of M402 + nabP + GEM versus nabP + GEM.
- Part A is ongoing, with several dose cohorts completed.
- Full Part A data are expected 1H 2014.

Phase 1/2 Study (cont.)

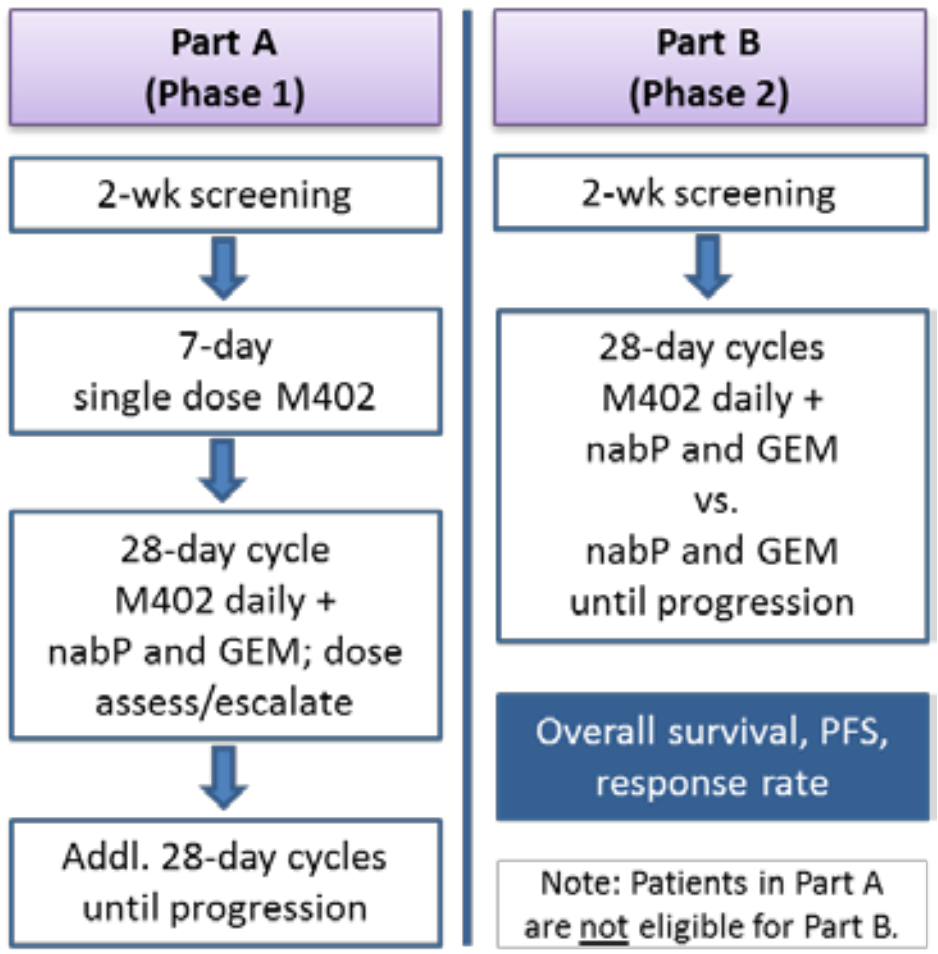
Part A
(Phase 1)
2-wk screening
7-day
single dose M402
28-day cycle
M402 daily +
nabP and GEM; dose
assess/escalate
Addl. 28-day cycles
until progression
Part B
(Phase 2)
2-wk screening
28-day cycles
M402 daily +
nabP and GEM
vs.
nabP and GEM
until progression
Overall survival, PFS,
response rate
Note: Patients in Part A
are not eligible for Part B.

Conclusions

- These and other promising data from in vitro and in vivo experiments support M402's evaluation in the ongoing clinical study.
- M402's broad mechanism of action may also support the future clinical investigation of M402 in a variety of other cancers.

Momenta Pharmaceuticals, Inc. , 675 W. Kendall St., Cambridge, MA – www.momentapharma.com

Poster presented at Cancer Advance, September 19, 2013; Harvard Medical School, Boston, MA